Antiretroviral Activity and Tolerability of Reverset (D-d4FC), a New Fluorocytidine Nucleoside Analog, When Used in Combination Therapy in Treatment-Experienced Patients: Results of Phase IIb Study RVT-203

C. Cohen,[1] C. Katlama,[2] R. Murphy,[3] J. Gathe,[4] C. Brinson,[5] G. Richmond,[6] P.-M. Girard,[7] J. Fessel,[8] A. Liappis,[9] E. Puglia,[10] B. Rodwick,[11] J. Nadler,[12] W. O'Brien,[13] K. Arasteh,[14] M. Otto,[15] S. Erickson-Viitanen,[16] and R. Levy[16]

[1]Community Research Initiative of New England, Boston, USA; [2]Hôpital de la Pitié, Paris, France; [3]Northwestern University, Chicago, USA; [4]Fannin Street, Houston, USA; [5]Central Texas Clinical Research, Austin, USA; [6]315 SE 14th Street, Fort Lauderdale, USA; [7]Hôpital St. Antione, Paris, France; [8]Kaiser Permanente, San Francisco, USA; [9]George Washington University Medical Center, Washington, DC, USA; [10]Research Center of Florida, Inc., Miami, USA; [11]Clinical Research of West Florida, Clearwater, USA; [12]Hillsborough County Health Department, Tampa, USA; [13]University of Texas Medical Branch, Galveston, USA; [14]EPIMED, Berlin, Germany; [15]Pharmasset, Inc. Tucker, USA; [16]Incyte Corp., Wilmington, USA

ABSTRACT

Objectives: Evaluate efficacy and tolerability of once daily doses of 50, 100 or 200 mg Reverset (RVT), a cytidine analog NRTI, in ARV-experienced patients.

Methods: Phase IIb, randomized, blinded, multinational, placebo-controlled 3 stage study consisting of: 2 wk add-on phase (RVT or placebo); 14-wk optimized treatment phase; and 8-wk safety phase (placebo pts allowed to cross over) in treatment-experienced patients (pts.).

Results: 199 pts enrolled at 25 sites; currently, 120 completed >16 wks of therapy. Mean baseline VL = 4.5 log10, presence of M184V: 60%, M41L: 60%, 4-6 TAMs: 50%, K65R: 6%. After 2 wks, mean VL changed by +0.007, -0.5, -0.3, and -0.7 log10 for placebo, 50, 100, and 200 mg groups respectively. Pts on 200 mg with 0-3 TAMs had 2-wk VL drop of 0.8 log10, with 4-6 TAMs a 0.6 log10 drop. Pts with M41L, M41L+L210W or w/o M41L showed similar VL decreases (0.7 log10). Pts with K65R or M184V + TAMs had more variable responses (mean 0.4 log10 drop); >20% pts had > 1 log10 drop. 33% pts didn't optimize at wk 2. Of these, current data shows stable VL decrease of ~0.6 log10 at wk 16 on 200 mg RVT. AEs generally mild, included headache, fatigue and GI disorders. In 35 subjects taking RVT with ddI, 12 (34%) had grade 4 hyperlipasemia, usually occurring after >12 wks of treatment. Among pts not on ddI, asymptomatic grade 4 lipase seen in 5.4% of pts on 200 mg RVT vs. 3.1% on placebo. Pancreatitis seen in 3 pts on RVT+ddI+TDF: 1 pt on ddI 250mg/TDF 450mg, 1 pt on ddI 400mg/TDF 300mg; all resolved off drugs.

Conclusions: RVT 200 mg is active in ARV-experienced pts and generally well tolerated. Because of the risk of elevated lipase and pancreatitis, RVT should not be used with ddI. Data support continued development of RVT.

Background Information

- **Preclinical studies demonstrated**
 - Active with viral mutants resistant to 3TC, TDF, AZT, other NRTIs, NNRTIs, and PIs
 - Long intracellular half-life (17 hours)
 - No mitochondrial toxicity or lactic acid increase (*in vitro*)
- **10-day add-on dosing demonstrated**
 - VL decrease of 0.8 logs with 200 mg qd dosing in therapy-experienced patients
 - VL decrease of 1.8 logs with 200 mg qd dosing in therapy-naïve patients

Chemical structure of **REVERSET™** (D-D4FC)

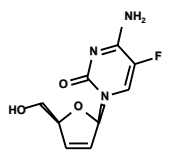
NH2
N
F
O
N
HO
O

β-D-2',3'-didehydro-2',3'-dideoxy-5-flurocytidine

RVT-203 Study Design

- Randomized, placebo-controlled, double-blind study in 3 phases:

2-week add-on	14-week treatment with optimized background regimen	8-week placebo cross-over

- Determine short-term potency/select dose
- Assess impact of resistance mutations
- Assess impact of concurrent ARVs

- Determine durability of response/select dose
- Assess development of resistance mutations
- Assess impact of optimized background
- Identify populations which will benefit most
- Assess longer term safety and tolerability

Baseline Characteristics

Parameter	Placebo (*N* = 47)	50 mg (*N* = 48)	100 mg (*N* = 49)	200 mg (*N* = 51)
Mean baseline VL, logs	4.52	4.25	4.40	4.59
No. with VL > 5 logs	4	7	11	14
No. who did not optimize at week 2	15	17	21	13
No. with 0–3 TAMS	25	28	27	29
No. with 4–6 TAMS	22	20	22	22
No. with both M41L and L210W	22 (47%)	20 (42%)	22 (45%)	25 (49%)
No. with M184V	31 (66%)	35 (73%)	30 (61%)	32 (63%)
No. with K65R	3	2	4	2

Demographics and Endpoints

Demographics

Parameter	*N* as of July 1, 2005
No. enrolled	199
No. completing week 16	171
No. completing week 24	112*
Percentage black or multi-racial	16.4%
Percentage female	8 %

*Study is still ongoing

Endpoints

- Primary variables
 - Mean change in viral load to week 2 and week 16
 - Adverse events (AEs)
- Secondary variables include
 - Proportions > 1 log decline
 - Patients who did/did not optimize at week 2

RESULTS

2-WEEK ADD-ON PHASE

Change in Viral Load From Baseline 2-Week Add-on Phase

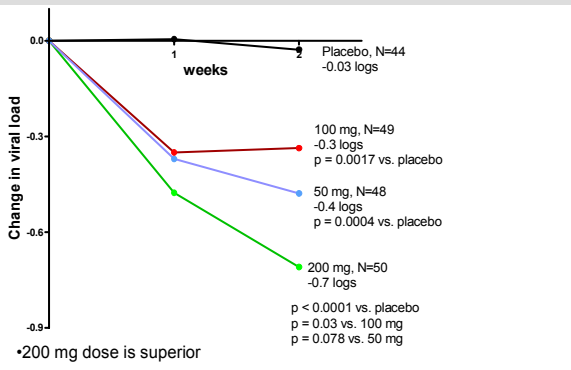
Change in viral load
weeks
0.0
-0.3
-0.6
-0.9
1
2
Placebo, N=44
-0.03 logs
100 mg, N=49
-0.3 logs
p = 0.0017 vs. placebo
50 mg, N=48
-0.4 logs
p = 0.0004 vs. placebo
200 mg, N=50
-0.7 logs
p < 0.0001 vs. placebo
p = 0.03 vs. 100 mg
p = 0.078 vs. 50 mg

•200 mg dose is superior

Effect of NRTIs in Baseline Regimen on 2-Week Viral Load Decline with 200 mg RVT

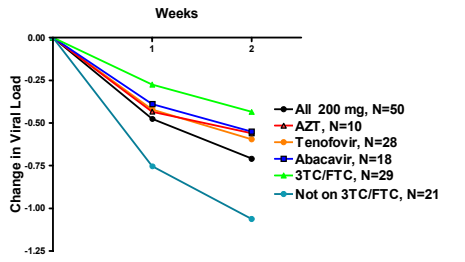
Weeks
Change in Viral Load
0.00
-0.25
-0.50
-0.75
-1.00
-1.25
1
2
All 200 mg, N=50
AZT, N=10
Tenofovir, N=28
Abacavir, N=18
3TC/FTC, N=29
Not on 3TC/FTC, N=21

- Activity retained with tenofovir, AZT, abacavir
- Activity decreased with 3TC/FTC
- Subjects not taking 3TC/FTC achieve > 1 log reduction in 2 weeks

Effect of RT Mutations on 2-Week Viral Load Decline with 200 mg RVT

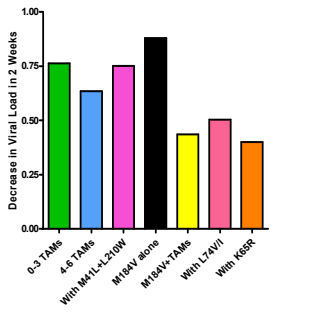
Decrease in Viral Load in 2 Weeks
1.00
0.75
0.50
0.25
0.00
0-3 TAMs
4-6 TAMs
With M41L+L210W
M184V alone
M184V+TAMs
With L74V/I
With K65R

- Activity similar with 0-3 or 4-6 TAMs
- Full activity with M41L + L210W mutation (with other TAMs)
- No loss in activity with M184V alone
- Somewhat diminished response with M184V+TAMs (but see week 16 result)
- > 0.5 log with L74 mutations
- 0.4 log with K65R

Note: Data for M184V alone and K65R represents pooled data from all doses because of small sample size

WEEK 16 (2-WEEK ADD-ON PHASE PLUS 14-WEEK OPTIMIZATION PHASE)

Study RVT-203: 16-week Efficacy Results

	Placebo N=40	RVT 50 mg N=42	RVT 100 mg N=42	RVT 200 mg N=43	200 mg vs Placebo
Decrease in VL (log_{10}) from BL: All	0.8	0.9	0.8	1.2	0.4
Non-optimizers	0.07	0.3	0.4	0.6	0.5
Responder Rate (% > 1 log_{10} decrease in VL): All	40%	38%	31%	54%	14%
Non-optimizers	8%	0%	11%	31%	23%

Sub-Group Analysis

Sub-Group	Δ VL		% > 1 log Decrease	
	Placebo	200 mg	Placebo	200 mg
Triple Class Failures N=47	-0.5	-0.9	26%	43%
No 3TC/FTC: All Subjects N=29	-0.5	-1.4	25%*	80%*
No 3TC/FTC: Non-Optimizers N=9	+0.3	-1.5	0%	80%

*p < 0.05

Presence of M184V Does Not Impact 16-Week Response to 200 mg RVT

Presence/absence of M184V with TAMs

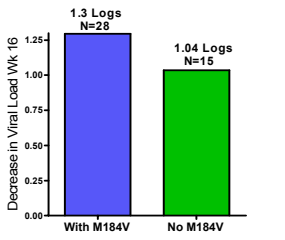
Decrease in Viral Load Wk 16
1.3 Logs
N=28
1.04 Logs
N=15
With M184V
No M184V

- Improved response without 3TC/FTC is not related to possible resistance to M184V+TAMs virus

RVT Provides Stable Reduction in Viral Load when Added to a Failing Regimen

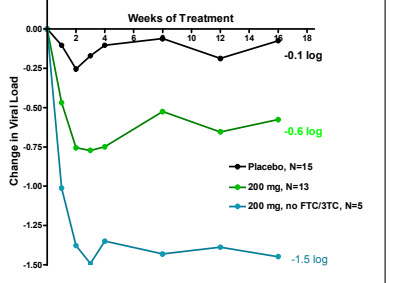
Weeks of Treatment
Change in Viral Load
-0.1 log
-0.6 log
-1.5 log
Placebo, N=15
200 mg, N=13
200 mg, no FTC/3TC, N=5

Results in subjects who did not optimize at week 2

Number of Subjects with Possibly Related Clinical Adverse Events of at Least Moderate Severity Through 16 Weeks

Event	Placebo N=47	RVT 50mg N=49	RVT 100 mg N=50	RVT 200 mg N=52
Nausea	0	2	1	1
Headache	0	1	1	2
Diarrhea	2	1	1	1
Pancreatitis	0	0	1	0
Myalgia	0	2	0	0
Abdominal Pain	0	0	1	0
Vomiting	0	0	0	1
Dry Mouth	0	0	1	0
Fatigue	0	0	0	1
Abnormal Feeling	0	1	0	0
Malaise	0	0	0	1
Fever	0	0	1	0
Gastroenteritis	0	0	0	1
Arthralgia	0	0	0	1
Muscle Cramp	0	0	1	0
Dizziness	0	0	0	1
Hypoaesthesia	0	0	0	1
Insomnia	0	0	1	0

Hyperlipasemia-Grade 4

Parameter	Placebo	50 mg	100 mg	200 mg
Patients not on ddI	1/39 3%	1/37 3%	2/34 6%	2/37 5%
Patients on ddI	0/8 0%	1/11 9%	4/15 27%	7/14 50%

- **Low frequency of hyperlipasemia without ddI**
- **High frequency of hyperlipasemia with ddI**
- **3 patients on 100 mg RVT had symptomatic pancreatitis**
- **RVT should not be used with ddI**

Pancreatitis

4 Cases of pancreatitis, all on 100 mg RVT

- **N=3 symptomatic pancreatitis**
 - One patient taking full dose (400 mg) ddI + tenofovir
 - One patient taking 250 mg ddI plus 1.5X dose tenofovir (450 mg)
 - One patient with ongoing alcohol abuse
- **N=1 asymptomatic, radiographic pancreatitis**
 - Taking recommended doses of ddI + tenofovir
- **All patients recovered**
 - Rapid, symptomatic and radiographic recovery
 - No complications
 - Lipase/amylase levels recovered more slowly

Other Possibly Related Serious Adverse Events in Subjects on RVT

- Anemia (1 patient on 200 mg RVT)
 - Also receiving AZT
- Neutropenia (1 patient on 200 mg RVT)
 - Worsening neutropenia when started on TMP-SMX for PCP
- Hypertriglyceridemia (1 patient on 100 mg RVT)
 - Developed worsening hypertriglyceridemia when started on fosamprenavir 1400 mg and ritonavir 200 mg once daily
- Pancytopenia (1 patient on 200 mg RVT)
 - Developed anemia (Grade 3), neutropenia (grade 3 and thrombocytopenia (Grade 2), resolved with discontinuation of study medication
 - No other evidence of bone marrow toxicity in the entire study

CONCLUSIONS

1. **200 mg Reverset provides antiviral suppression in highly ARV-experienced patients**
 - 2-week add-on phase: -0.7 logs; -1.1 log_{10} without 3TC/FTC
 - 16 weeks: 54% response overall (> 1.0 logs); 80% without 3TC/FTC
2. **Reverset is generally well tolerated**
 - Asymptomatic hyperlipasemia when used with ddI
 - RVT should not be used with ddI
3. **Reverset activity retained with key mutations**
 - To date, no novel mutations identified in RVT-exposed patients
 - No K65R emerged to date in RVT-exposed patients
4. **Reverset warrants further development**
 - Phase III trial planning in progress

ACKNOWLEDGMENTS

Participating subjects

Investigators

United States
Calvin Cohen, Joseph Gathe, Jeffrey Fessel, Angelike Liappis, Barry Rodwick, William O'Brien, Jeffrey Galpin, Donna Mildvan, Corklin Steinhart, Rob Murphy, Cynthia Brinson, Gary Richmond, Edgardo Puglia, Jeffrey Nadler, Steven O'Marro, George Beatty, Kathleen Squires, Gerald Pierone

France
Christine Katlama, Pierre-Marie Girard

Germany
Keikawus Arasteh, Dirk Schurmann, Shlomo Staszewski, Hans-Juergen Stellbrink, Albrecht Stoehr

Sponsors
Incyte Corporation, Pharmasset, Inc.