P19.5/05

Analysis of the cost of full virological suppression for highly treatment-experienced, HIV-infected patients in the POWER trials in different European healthcare settings

Hill AM,[1] Clotet B,[2] Johnson M,[3] Stoll M,[4] Smets E[5]

[1]University of Liverpool, Liverpool, UK and Tibotec BVBA, Mechelen, Belgium; [2]Hospital Universitari Germans Trias i Pujol and irsiCaixa Foundation, Barcelona, Spain; [3]Royal Free Hospital, London, UK; [4]Medical University of Hannover, Hannover, Germany; [5]Johnson & Johnson Pharmaceutical Services, Mechelen, Belgium

Address correspondence to: Andrew Hill, PhD, Pharmacology Research Laboratories, University of Liverpool, 70 Pembroke Place, Liverpool, L69 3GF, UK; Fax +44 (0) 151 794 5656; E-mail: microhaart@aol.com

Introduction

- HIV RNA suppression <50 copies/mL is an accepted surrogate marker for progression risks in clinical studies of antiretroviral therapy (ART).[1] Economic analyses normally assess cost per quality adjusted life-year saved, but these have not been standardised in HIV. A more direct assessment of economic value is the total cost of ART relative to rates of full HIV RNA suppression.[2]
- This analysis used data from the POWER 1 and 2 clinical trials[3] to assess treatment costs associated with the use of darunavir with low-dose ritonavir (DRV/r) versus investigator-selected control PI (CPI) in highly pretreated HIV-infected adults. The new methods were applied using ART costs in 13 European countries, to test the sensitivity of the conclusions in different healthcare settings.

Methods

- The data from the POWER trials included 131 patients randomised to DRV/r 600/100mg bid, and 124 patients receiving CPI. Summary of baseline and 48-week virological efficacy data (% with HIV RNA <50 copies/mL) data are shown in Table 1.

Table 1. Baseline demographics of the POWER 1 and 2 trials.

Treatment arm	DRV/r (N=131)	CPI(s) (N=124)
Baseline characteristics		
Mean age (years)	44	44
Male (%)	89	88
Caucasian (%)	81	73
Mean HIV RNA (log_{10})	4.6	4.5
Median CD4 cell count (cells/mm^3)	153	163
CDC Class C (%)	36	43
Prior ARV therapy (%)		
≥4 NRTI	93	97
≥1 NNRTI	96	97
≥2 PI	94	99
Prior enfuvirtide	18	16
HIV RNA <50 copies/mL at Week 48 (%)		
Total population	45	10
Enfuvirtide naïve use	58	11
No enfuvirtide use	44	10
≤1 active drug in OBR	37	2
≥2 active drugs in OBR	56	17

OBR = optimised background regimen

- Data on the actual usage of ART in POWER 1 and 2 were used to calculate the total annual cost of treatment for the DRV/r 600/100mg bid and CPI treatment arms (Table 2). Patients in the CPI arm used either one or two PIs boosted by ritonavir. Twenty-seven per cent of patients in the CPI arm used a dual-boosted PI.

Table 2. ARV drug use by treatment arm in the POWER 1 and 2 trials.

Treatment arm	Dose	DRV/r (%)	CPI(s) (%)
Nucleoside analogues			
Lamivudine	150mg bid or 300mg qd	52	57
Emtricitabine	200mg qd	21	16
Zidovudine	300mg bid	17	27
Didanosine	400mg qd	37	37
Tenofovir	300mg qd	84	86
Stavudine	40mg bid	15	15
Abacavir	300mg bid	15	23
Protease inhibitors			
Atazanavir/r	300/100mg qd	0	17
Fosamprenavir/r	700/100mg bid	0	34
Indinavir/r	800/100mg bid	0	2
Saquinavir/r	1,000/100mg bid	0	35
LPV/r	400/100mg bid	0	36
Nelfinavir	1,250mg bid	0	1
DRV/r	600/100mg bid	100	0
Fusion inhibitor			
Enfuvirtide	90mg bid injection	47	42

LPV/r = lopinavir/ritonavir

- Costs were calculated by country, assuming that patients continued to take all treatments assigned at baseline for a full 52 weeks. Country-specific costs of antiretrovirals (ARVs) were taken from published sources in July 2007. Currencies in Europe were converted to Euros at July 2007 exchange rates.
- In addition, the following sensitivity analyses were conducted:
 1. The CPI was assumed to be exclusively LPV/r. The costs of LPV/r were included in the CPI arm. Because patients treated with LPV/r showed virological efficacy consistent with the other patients in the CPI arm,[4] the same efficacy was assumed for LPV/r in this sensitivity analysis. This analysis lowers the cost of the CPI arm, as 27% of the patients received double-boosted PI regimens in this arm
 2. The analysis was repeated for the subset of patients who a) took enfuvirtide for the first time ('naïve users') in the trials and b) did not receive enfuvirtide during the trial, separately. This analysis was performed because of the high costs of enfuvirtide relative to other ARVs, and its apparent contribution to the efficacy of the DRV/r arm
 3. Finally, the analysis was also repeated for the subset of patients who a) had ≤1 active drug in the OBR and b) had ≥2 active drugs in the OBR. The latter subset of patients showed higher overall efficacy in both the DRV/r and CPI arms (Table 1).
- The incremental cost-efficacy ratios from the POWER trials were also compared with results from recently conducted clinical trials in treatment-experienced patients with a profile comparable to that of the POWER participants: the RESIST trials of tipranavir/r (TPV/r) and the TORO trials of enfuvirtide.[5,6]

Results

- In this analysis, we applied a method of calculating costs per effective treatment response, defined as HIV RNA <50 copies/mL to the 48-week results of the pooled POWER 1 and 2 trials of DRV/r versus CPI in highly treatment-experienced patients.
- In these trials, based on medication acquisition costs only, the mean per-patient cost of a year's therapy with DRV/r-based ART was between 1.4–20% higher than the cost of CPI-based treatment, dependent on the European healthcare setting considered.
- However, significantly more patients reached undetectable viral load (HIV RNA <50 copies/mL) with a regimen containing DRV/r (45%) than with CPI (10%).
- Consequently, DRV/r therapy generated a 73–76% lower cost per treatment response than CPI therapy in this analysis, in terms of the cost per patient with HIV RNA suppression to <50 copies/mL at Week 48 (Table 3; Figure 1).

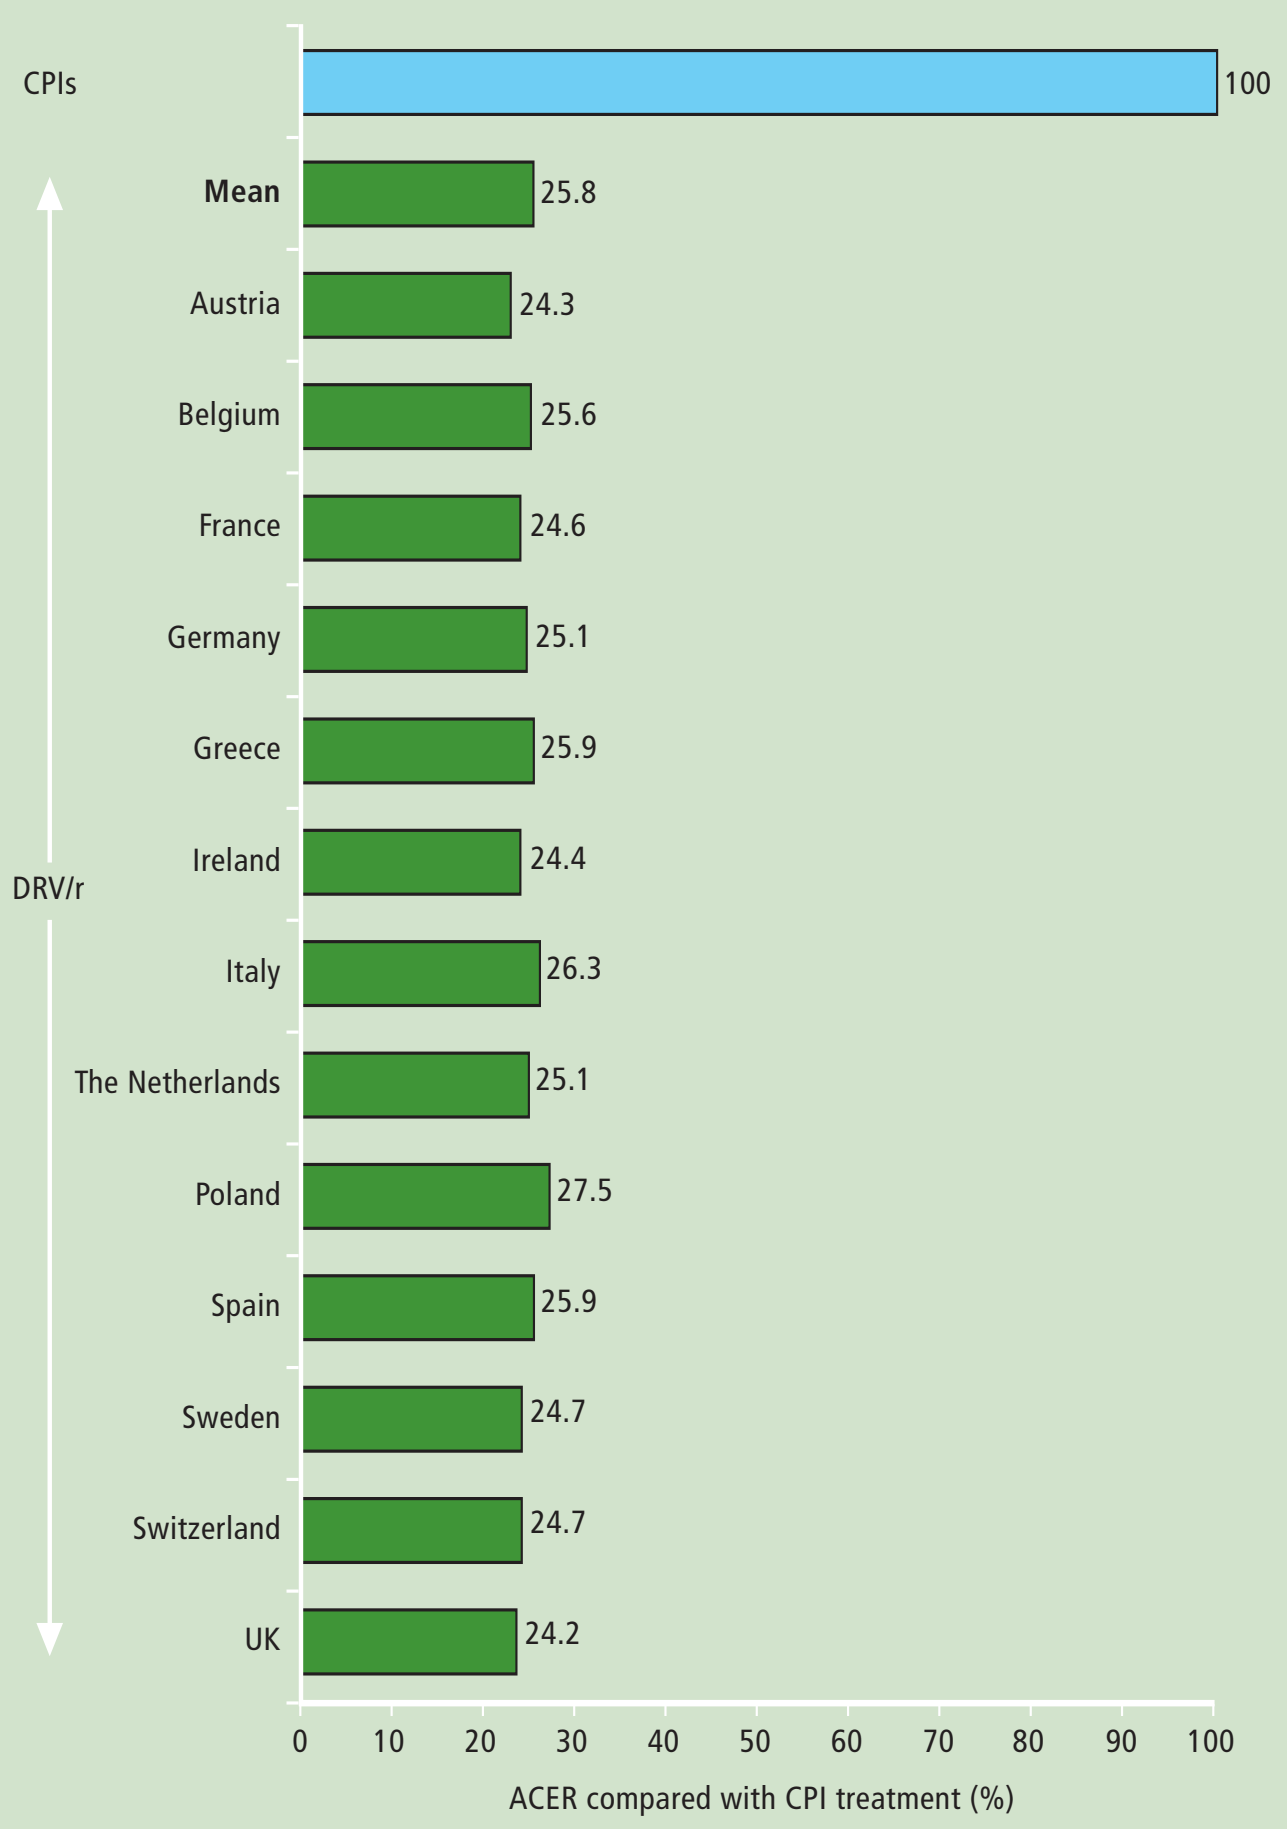

CPIs
100
DRV/r
Mean
25.8
Austria
24.3
Belgium
25.6
France
24.6
Germany
25.1
Greece
25.9
Ireland
24.4
Italy
26.3
The Netherlands
25.1
Poland
27.5
Spain
25.9
Sweden
24.7
Switzerland
24.7
UK
24.2
0 10 20 30 40 50 60 70 80 90 100
ACER compared with CPI treatment (%)

ACER = actual cost efficacy ratio (the cost per patient with HIV RNA levels <50 copies/mL at Week 48)

Figure 1. Reduction in the actual cost efficacy ratio (ACER) with DRV/r compared with use of CPIs in European countries.

Table 3. Annual mean per-patient costs of treatment, cost per patient with HIV RNA <50 copies/mL in European countries.

Country	DRV/r arm Mean cost (€)	DRV/r arm ACER	CPI arm Mean cost (€)	CPI arm ACER	Per cent reduction in ACER	ICER
Austria	25,944	57,653	23,386	233,858	–75.7	7,308
Belgium	28,205	62,677	24,465	244,650	–74.4	10,686
France	23,694	52,653	21,406	214,063	–75.4	6,537
Germany	26,842	59,649	23,799	237,990	–74.9	8,694
Greece	21,777	48,393	18,664	186,640	–74.1	8,894
Ireland	30,217	67,149	27,563	275,630	–75.6	7,583
Italy	21,741	48,313	18,349	183,490	–73.7	9,691
The Netherlands	26,805	59,567	23,717	237,170	–74.9	8,823
Poland	23,150	51,444	18,687	186,870	–72.5	12,751
Spain	23,055	51,233	19,749	197,490	–74.1	9,446
Sweden	26,819	59,598	24,156	241,560	–75.3	7,609
Switzerland	28,915	64,257	26,078	260,780	–75.3	8,106
UK	26,643	59,207	24,465	244,650	–75.8	6,223
Mean	**25,677**	**63,813**	**22,653**	**226,526**	**–74.2**	**8,642**

ACER = actual cost efficacy ratio (the cost per patient with HIV RNA levels <50 copies/mL at Week 48)
ICER = incremental cost efficacy ratio (the additional cost per extra patient with HIV RNA levels <50 copies/mL)

- The lower cost per treatment response for DRV/r versus CPI therapy was confirmed in each of the 13 European countries included in this analysis, in sensitivity analyses controlling for enfuvirtide use and the number of active drugs (Table 4).

Table 4. Sensitivity analysis for UK costs in euros.

Sensitivity analysis for UK costs	HIV RNA <50 copies/mL (%) DRV/r	HIV RNA <50 copies/mL (%) CPI	Cost (€) DRV/r	Cost (€) CPI	ACER DRV/r	ACER CPI
Analysis						
Base case	45	10	27,275	24,860	60,611	248,596
CPI is LPV/r only	45	10	27,275	23,337	60,611	233,366
Enfuvirtide used for all patients	58	11	37,752	37,488	65,089	340,803
No enfuvirtide used	44	10	17,954	15,956	40,804	159,559
≤1 active drug in OBR	37	2	27,275	24,860	73,716	1,242,978
≥2 or more active drugs in OBR	56	17	27,275	24,860	48,705	146,233

ACER = actual cost efficacy ratio (the cost per patient with HIV RNA levels <50 copies/mL at Week 48)

- Finally, the incremental cost-efficacy ratios of DRV/r versus CPIs were consistently lower in comparison with equivalent analyses for TPV/r (RESIST trials) and enfuvirtide (TORO trials) in all European countries considered in this analysis (Table 5).

Table 5. Incremental efficacy, costs and cost-efficacy for DRV versus other ARVs for treatment-experienced patients: UK prices in euros (48-week data).

Trial	Treatment comparison	Additional patients reaching HIV RNA <50 copies/mL (%)	Cost difference (€)	ICER
TORO 1 and 2	Enfuvirtide vs OBR	10 (18 vs 8)	20,386	203,855
RESIST 1 and 2	TPV/r vs CPI/r	13 (23 vs 10)	6,191	47,620
POWER 1 and 2	DRV/r vs CPI/r	35 (45 vs 10)	2,149	6,139

ICER = incremental cost efficacy ratio (the additional cost per extra patient with HIV RNA levels <50 copies/mL)

Conclusions

- The cost of ART per patient with full HIV RNA suppression was 73–77% lower in the DRV/r 600/100mg bid arm of the POWER 1 and 2 trials, relative to the CPI arm. This finding was consistent across Europe.
- This economic advantage was maintained in sensitivity analyses, adjusting for the type of CPI, use of enfuvirtide, and the number of active drugs in the OBR.
- The (incremental) cost per patient with an undetectable viral load is a valuable, useful and relevant economic measure, in particular for HIV care providers and payers managing ARV medication budgets, and should become a key element of the assessment of the economic value of ARV therapy and other HIV-related interventions, as part of a set of diverse, complementary economic analyses.

References

1. Hammer SM, et al. JAMA 2006;296:827–43.
2. Basu RP, et al. HIV Clin Trials 2006;7:309–18.
3. Clotet B, et al. Lancet 2007;369:1169–78.
4. Lefebvre E, et al. 8th International Congress on Drug Therapy in HIV Infection, Glasgow, UK, 12–16 November 2006. Abstract P29.
5. Hicks CB, et al. Lancet 2006;368:466–75.
6. Nelson M, et al. J Acquir Immune Defic Syndr 2005;40:404–12.

Supported by Tibotec

Presented at the 11th European AIDS Conference, Madrid, Spain, 24–27 October 2007.

This poster is available on-line at www.tibotec.com.