P18.1/01

A Prospective Epidemiological Study to Determine the Prevalence of *HLA-B*5701* in French HIV-1 Infected Patients: PEPI Study

JM Molina[1], G Pialoux[2], F Raffi[3], G Force[4], PM Girard[5], R Verdon[6], P Dellamonica[7], M Partisani[8], G Nachbaur[9], C Roché[9], H Pearce[10]

[1]Hôpital St Louis, Service des Maladies Infectieuses, Paris, France; [2]Hôpital Tenon, Service des Maladies Infectieuses, Paris, France; [3]Hôpital Hotel Dieu, Service des Maladies Infectieuses, Nantes, France; [4]Hôpital Notre Dame du Bon Secours, Service des Maladies Infectieuses, Levallois-Perret, France; [5]Hôpital St Antoine, Service des Maladies Infectieuses, Paris, France; [6]CHU de la Côte de Nacre, Service des Maladies Infectieuses, Caen, France; [7]Hôpital de l'Archet, Service Infectiologie, Nice, France; [8]Hôpitaux Universitaires de Strasbourg, CISIH, Strasbourg, France; [9]Laboratoire GlaxoSmithKline, Marly le Roi, France, [10]GlaxoSmithKline, Brentford, United Kingdom

Introduction

Carriage of the human leucocyte antigen allele *HLA-B*5701* has been strongly linked to the occurrence of abacavir (ABC) hypersensitivity reaction (HSR) in Caucasian populations[1,2], and data from the SHAPE study has confirmed a similar strength of association in non-Caucasians (African-Americans)[3]. The large, blinded PREDICT-1 study recently showed the utility of prospective screening for *HLA-B*5701* by demonstrating a dramatic reduction in the incidence of ABC HSR when ABC-containing therapy was avoided in ABC-naïve patients testing positive for *HLA-B*5701*, relative to a contemporary control population who were not screened before treatment.[4]

*HLA-B*5701* carriage prevalence varies between populations and geographical regions,[5] and to date, there are limited epidemiological data on the prevalence of *HLA-B*5701* in HIV-1 infected subjects as opposed to the general population. There are also limited data on the distribution of *HLA-B*5701* carriage across the various European regions and ethnotypes.

Objective

Primary Objective

- To determine the prevalence of *HLA-B*5701* in the French HIV-1 infected population.

Secondary Objectives

- To assess the prevalence of *HLA-B*5701* in the major French ethnotypes in the study population.
- To describe *HLA-B*5701* results provided by local methods with respect to those obtained from a central provider.

Methods

Study Design

- This was a cross-sectional observational study to evaluate the prevalence of *HLA-B*5701* in the major French ethnotypes according to the FDA and ICH guidelines.
- Any HIV-1 infected adult subject was eligible for this study. All subjects were to be consented prior to any study specific procedure.
- Subjects provided tissue samples (cheek cells and blood), which were used to assess *HLA-B*5701* status by central and local methodologies.
- The study was proposed to as many hospital centres as possible.
- Within each centre, patients were included consecutively when they came for their clinic visit, with respect of a targeted inclusion figure that was given to each centre (calculated as a subset of the whole patient population of the centre).
- Each investigator was asked to register the ethnicity of each patient coming to their visit during the inclusion period. This registry was used to assess how representative the sample was within each centre and to calculate correction factors if necessary. If ever the proportions in the different ethnic groups were not respected at the end of the study on the national population (as assessed with the investigators registry), the weight of the included patients within a centre and within an ethnic group were to be included in the analysis (sensitivity analysis taking into account these corrections of sampling was anyway planned in order to assess the consistency of the estimations).

Study Population

- The per-protocol population was the primary population for the calculation of *HLA-B*5701* prevalence according to central and local results.
- The 'registry population' consisted in all subjects recorded in the registry, included or not included in the study. The registry population was used to describe subjects' ethnicity in order to establish representativeness of per-protocol population.
- Safety has been assessed on the population of subjects included in the study.

Statistical Methods

- Assuming an expected prevalence of 6% to 8% in the 'white' sub group (origins in any of the original peoples of Europe or in any of the original peoples of Middle East or North Africa) and of 2% in the 'black' subgroup (origins in any of the black racial groups of Africa), the emphasis was put on the precision of the 'white' subgroup. With a precision of 1.25% for a prevalence of 6%, 1086 subjects should be enrolled.
- Considering the distribution of the ethnic groups in the French population, to reach this target of 1086 'white' HIV-1 infected subjects, a total of 1975 subjects should be included.
- With these figures, it was anticipated that the prevalence in the 'black' population would be obtained with almost a 1% precision of the 95%CI.
- To take into account attrition (non-evaluable, sample or missing data regarding ethnicity for instance) an attrition rate of 20% was expected and 2469 patients were to be included in the study.
- In all, 2371 subjects have been included in the study and 4142 were notified in the registry between 13th March and 31st May 2007 by 86 centers.
- 21 subjects did not meet criteria to be involved in the Per Protocol analysis (19 did not have results for the *HLA-B*5701* test and 2 presented with a deviation to inclusion criteria).

Results

Table 1. Baseline Characteristics (Per Protocol Population)

	N = 2350 patients
Females, n (%)	698 (30)
Median age, years (range)	43 (18-79)
Geographic ancestry*, n (%)	
African/African Heritage	523 (22%)
American Indian or Alaska Native	16 (<1%)
Asian-Central/south Asian Heritage	19 (<1%)
Asian-East Asian Heritage	4 (<1%)
Asian-Japanese Heritage	2 (<1%)
Asian-South East Asia Heritage	22 (<1%)
Native Hawaiian or Other Pacific Islander	3 (<1%)
White-Arabic/north African Heritage	164 (7%)
White-White/Caucasian European Heritage	1657 (71%)

* One patient could be classified in ≥1 geographic ancestry category.

Figure 1. Main Personal Geographic Ancestries Associations

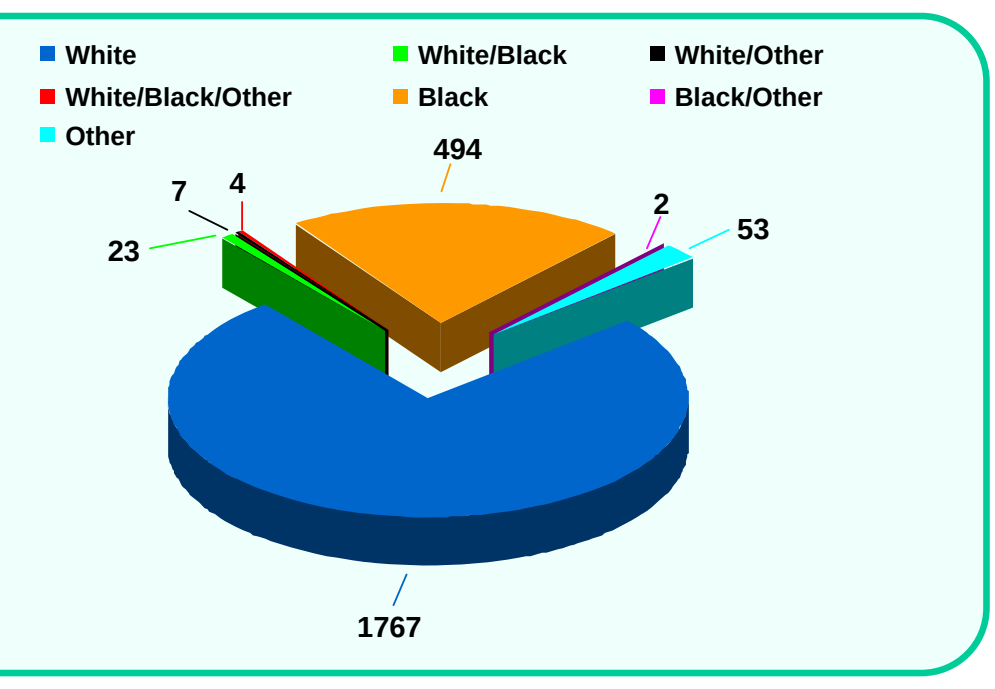
White
White/Black
White/Other
White/Black/Other
Black
Black/Other
Other
494
7
4
23
2
53
1767

Figure 2. Summary of Geographic According to Geographic Ancestry of Biological Relatives (Mother and Father)

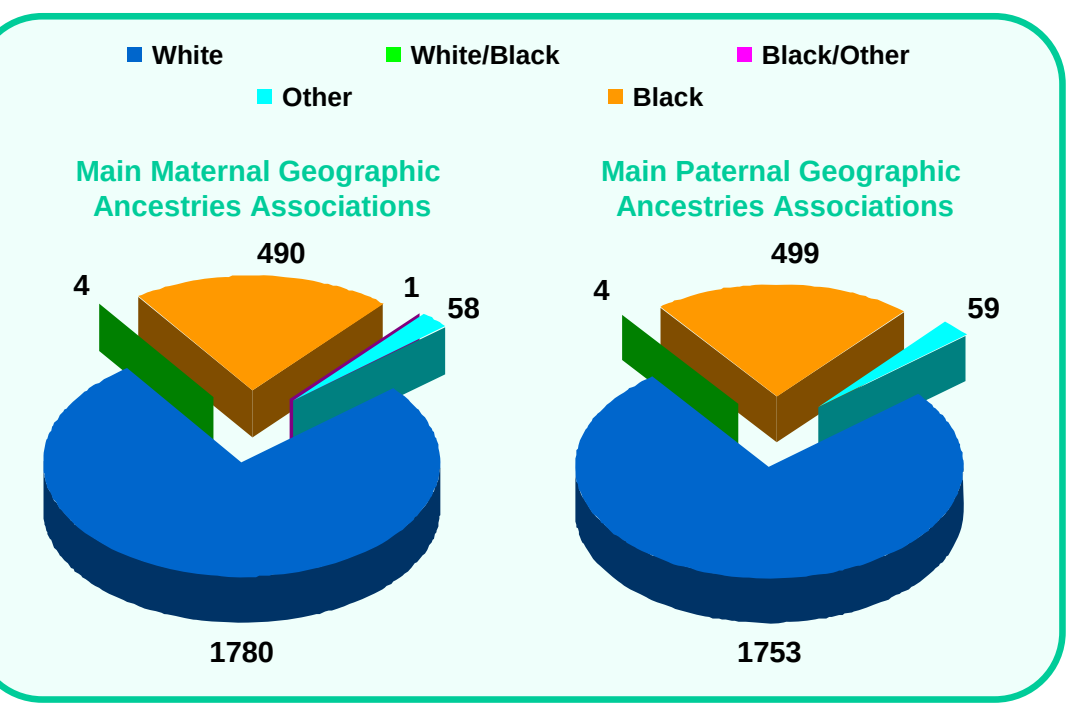
White
White/Black
Black/Other
Other
Black
Main Maternal Geographic Ancestries Associations
Main Paternal Geographic Ancestries Associations
490
4
1
58
1780
499
4
59
1753

- When compared to the Registry Population, there was no difference observed in the breakdown of geographic ancestries and no sensitivity analysis was performed.

Table 2. *HLA-B*5701* Prevalence According to Central Assessment (Buccal Swab) and Local Screening Methods (Blood)

	Central Laboratory	Local Laboratories
Total population (%) **[95% CI] (%)** ***(n/N)***	5.32 **[4.48 – 6.30]** ***(125/2350)***	5.22 **[4.38 – 6.20]** ***(120/2300)***
White* (%) **[95% CI] (%)** ***(n/N)***	6.90† **[5.81 – 8.18]** ***(122/1767)***	6.76 **[5.67 – 8.04]** ***(117/1731)***
Black* (%) **[95% CI] (%)** ***(n/N)***	0.41 **[0.11 – 1.46]** ***(2/494)***	0.42 **[0.11 – 1.50]** ***(2/481)***
Other*‡ (%) **[95% CI] (%)** ***(n/N)***	1.89 **[0.33 – 9.94]** ***(1/53)***	1.92 **[0.34 – 10.12]** ***(1/52)***

*Only patients with a single ethnicity are shown
†Within the White subgroup, prevalence, as assessed by central laboratory results, was 7.06% [5.92 – 8.40%] (117/1657) for patients of European origin and 6.10% [3.35 – 10.86%](10/164) for patients of Middle Eastern or North African origin.
‡Asian or other origin

Figure 3. *HLA-B*5701* Prevalence According to Central and Local Results

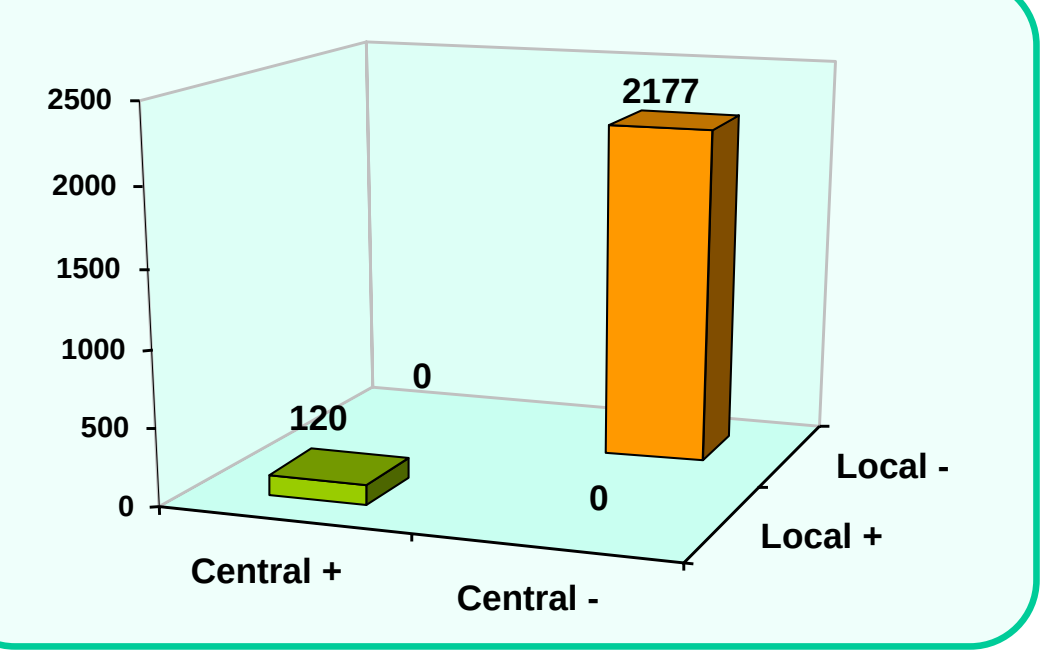
2500
2000
1500
1000
500
0
2177
120
0
0
Local -
Local +
Central +
Central -

- Concordance between central and local laboratory results was 100%.
- The analysis taking into account the multiple answers regarding ethnicity did not show any modification in the estimation of the prevalence in different subgroups.
- No safety issue was reported during the course of the study.

Conclusions

- **In the French HIV infected population the overall prevalence of *HLA-B*5701* was 5.32% according to central laboratory results.**
- **The prevalence according to major ethnotypes were 6.90% in the white subgroup (7.06% in the original peoples of Europe and 6.10% in the original peoples of Middle East or North Africa), 0.41% in the black subgroup and 1.89% in the other ethnicities subgroup.**
- **There was 100% concordance between *HLA-B*5701* results obtained from local testing laboratories and the central provider.**

Acknowledgements

The authors wish to thank all of the study participants, investigators, coordinator and the GSK PEPI Study Team for their contributions.

References

1. Mallal S *et al. Lancet* 2002; 359:727–732.
2. Hetherington S *et al. Lancet* 2002; 359:1121–1122
3. Saag M *et al.* 4th IAS Conference, Sydney, Australia 2007. Abstract WEAB305
4. Mallal S *et al.* 4th IAS Conference, Sydney, Australia 2007. Abstract WESS101
5. Nolan D *et al. J HIV Ther* 2003; 8:36–41

11th European AIDS Conference, 24–27 October 2007, Madrid, Spain