9329

Cost-effectiveness of HLA-B*5701 Testing Prior to Initiation of Abacavir Sulfate

Farkouh R,[1] Earnshaw SR,[1] Kauf TL,[2] Watson ME,[3] Maroudas P[4]

[1]RTI Health Solutions, Research Triangle Park, NC, [2]University of Florida, Gainesville, FL

[3]GlaxoSmithKline, Research Triangle Park, NC, [4]GlaxoSmithKline, Stockley Park, UK

Introduction

- Prospective genetic testing for the HLA-B*5701 allele is a proven strategy for reducing the incidence of abacavir (ABC) hypersensitivity reaction (HSR).[1]
- Current United States (US) treatment guidelines recommend testing for HLA-B*5701 before starting patients on an ABC-containing regimen. However, when HLA-B*5701 testing isn't readily available, it is reasonable to initiate an ABC-containing regimen with appropriate clinical counseling and monitoring for signs of HSR.
- This research prospective features HLA-B*5701 testing evaluated in a real-world, clinical practice setting by utilizing data from published open-label studies as compared to the blinded clinical trial protocol utilized in PREDICT-1, thus capturing more of the benefits of prospective testing.

Methods

We combined an HLA-B*5701 cost-effectiveness model with the Antiretroviral Drug Valuation and Cost-Effectiveness (ADVANCE) simulation model[2] to estimate the cost-effectiveness of initiating highly active antiretroviral therapy (HAART) with fixed-dose ABC plus lamivudine (3TC) compared to fixed-dose tenofovir (TDF) plus emtricitabine (FTC) from the perspective of the health care system. The analysis considered ABC-induced HSR (captured in the HLA-B*5701 cost-effectiveness model) and TDF-induced renal failure (captured in the ADVANCE model).

The HLA-B*5701 Cost-effectiveness Model

- A decision-analytic model (Figure 1) was used to evaluate costs and outcomes during the first 60 days of HAART where ABC use is informed by prospective HLA-B*5701 testing.
 - Patients entered the model when the decision to initiate HAART was made.
 - Comparators in the model included the following:
 - Fixed-dose ABC 600 mg/3TC 300 mg + efavirenz (EFV) 600 mg, conditional on results of prospective HLA-B*5701 testing;
 - HLA*B-5701-negative patients received ABC/3TC + EFV.
 - HLA*B-5701-positive patients and patients who tested negative but were diagnosed with clinically-suspected HSR received TDF/FTC + EFV.
 - Unconditional initiation of TDF 300 mg with FTC 200 mg + EFV 600 mg.
 - HLA*B-5701-negative patients on ABC/3TC + EFV may experience HSR-like symptoms, referred to in the model as "patient-suspected HSR."
 - Patient-suspected HSRs were managed with either a physician's office or emergency department visit.
 - The patient's symptoms were assessed and declared as "clinically-diagnosed HSR" or not HSR. They later continued with ABC/3TC + EFV, while the former ceased ABC immediately.

Figure 1. Cost-effectiveness Model Structure

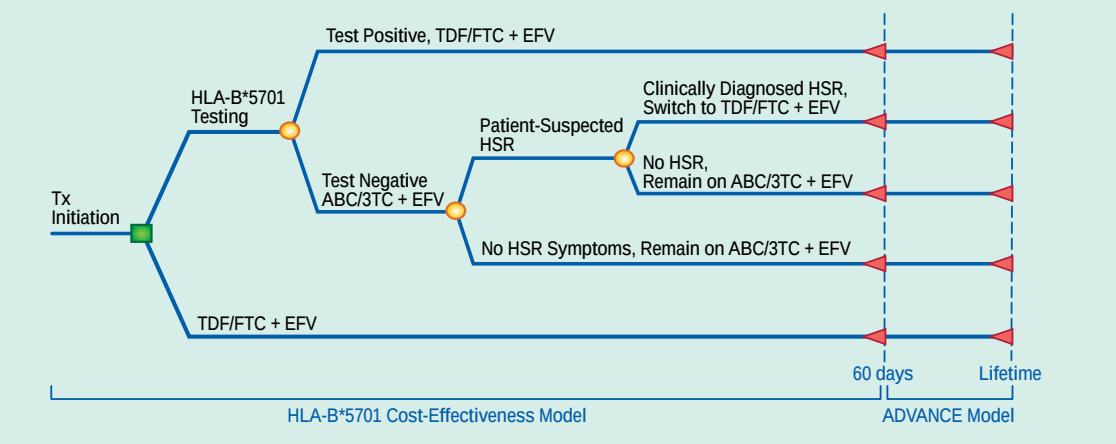
Test Positive, TDF/FTC + EFV
HLA-B*5701 Testing
Clinically Diagnosed HSR, Switch to TDF/FTC + EFV
Patient-Suspected HSR
No HSR, Remain on ABC/3TC + EFV
Test Negative ABC/3TC + EFV
Tx Initiation
No HSR Symptoms, Remain on ABC/3TC + EFV
TDF/FTC + EFV
60 days
Lifetime
HLA-B*5701 Cost-Effectiveness Model
ADVANCE Model

- Table 1 presents the inputs to the HLA-B*5701 cost-effectiveness model.

Table 1. Input Parameter Values and Sensitivity Analysis Ranges

Parameter	Base-Case Value	Lower Bound	Upper Bound	Source/Assumption
Drug costs				
ABC/3TC	$868.30	$694.64	$1,041.96	Red Book, 2007[3] AWP
TDF/FTC	$934.50	$747.60	$1,121.40	
EFV	$531.04	$424.83	$637.25	
LPV/r	$794.99	$642.62	$963.94	
HSR cost: patient suspected	$166.14	$0.00	$419.65	Table 2
HSR cost: clinically suspected	$799.85	$171.72	$1,739.12	Table 2
HLA-B*5701 test cost	$87.92	$50.00	$150.00	LabCorp, 2007[4]
60-day renal monitoring costs	$0.00	$0.00	$160.00	US Department of Health and Human Services, 2006[5]; RTI-HS, 2007[6]
HLA-B*5701 test negative predictive value	99.44%	91.80%	99.44%	Aggregate NPV based on four non-blinded trials[7–10]
HLA-B*5701 prevalence	5.57%	5.47%	10.00%	Mallal et al., 2008[1]
Rate of HSR symptoms among patients who test negative for the allele and initiate ABC	3.00%	0.00%	5.00%	RTI-HS, 2007[6]
HSR incidence (no-test arm)	6.00%	5.00%	6.10%	Epzicom, 2007[11]
Proportion of false HSRs in the no-test arm	50%	25%	75%	RTI-HS, 2007[6]
HIV + baseline utility	0.829	0.75	0.90	Kauf et al., 2008[12]
HSR utility decrement	–0.033	–0.10	0.00	Kauf et al., 2008[12]
Onset of HSR (days)	9	5	13	Mallal et al., 2008[1]
Wash-out period (days)	7	0	14	RTI-HS, 2007[6]

AWP = average wholesale price; LPV/r = ritonavir-boosted lopinavir.

- HSR costs were calculated by combining health care resource use estimates provided by infectious disease specialists with unit costs obtained from the Resource Based Relative Value Scale (RBRVS), database analyses, and published literature. Details of these calculations appear in Table 2.

Table 2. Cost Calculation for Patient-Suspected and Clinically Diagnosed Hypersensitivity Reactions

Resources Utilized	Unit Cost	No. of Units	Patient Suspected: Percentage of Patients Receiving Treatment	Patient Suspected: Costs	Clinically Diagnosed: Percentage of Patients Receiving Treatment	Clinically Diagnosed: Costs	Source/ Assumptions
Initial phone call (patient to doctor)	$37.14	1	100.0%	$37.14	100.0%	$37.14	RTI-HS, 2007[6] Ingenix, 2007[13]
Outpatient physician's appointment/ clinic visit	$134.58	1	67.0%	$90.17	67.0%	$90.17	RTI-HS, 2007[6] RTI-HS, 2007[14]
Emergency department visit	$608.68	1	6.2%	$37.74	6.2%	$37.74	RTI-HS, 2007[6] RTI-HS , 2007[14]
Inpatient general ward (1 day)	$2,167.10	4	0.00%	$0.00	5.3%	$459.42	RTI-HS, 2007[6] RTI-HS , 2007[14]
Acetaminophen	$7.15	1	10.0%	$0.72	10.0%	$0.72	NDC: 50580-0496-60[3]
Ibuprofen	$3.73	1	10.0%	$0.37	10.0%	$0.37	NDC: 19458-9558-02[3]

NDC = National Drug Code.

- The real-world effectiveness of HLA-B*5701 testing was taken from four published studies where HLA-B*5701 testing was evaluated in a non-blinded setting.[7–10]
- Incremental lifetime costs, life-years, quality-adjusted life-years (QALYs), and the incremental cost per QALY gained were then calculated.
- Multiple one-way sensitivity analyses were performed to assess parameter uncertainty.

The ADVANCE Model

- The ADVANCE model was used to derive long-term costs and outcomes beyond the initial 60-day treatment period.
- Lifetime treatment pathways were defined by five regimens (Table 3); patients transitioned to the next regimen following virologic failure.

Table 3. Treatment Pathways[a]

Treatment Pathways	ABC/3TC + EFV With Prospective Testing	TDF/FTC + EFV
First-line regimen	ABC/3TC + EFV	TDF/FTC + EFV
Second-line regimen	TDF/FTC+ LPV/r	3TC/ZDV + LPV/r
Three-class–experienced regimen[b]	TCE	TCE
Salvage regimen 1	ATZ/r + TDF + 3TC	ATZ/r + TDF + 3TC
Salvage regimen 2	ATZ/r + ZDV + 3TC + T20	ATZ/r + ZDV + 3TC + T20

T20 = enfuvirtide; TCE = three-class–experienced; ZDV = zidovudine.

[a] The ADVANCE model ignores all HAART-induced adverse events, with the exception of TDF-induced acute renal failure. ABC-induced HSR is captured within the HLA-B*5701 cost-effectiveness model.

[b] Three-class–experienced regimen is assumed to have similar regimen attributes as salvage regimen 1.

- Drug costs (Table 1) and efficacy estimates (Table 4) were used as inputs to the ADVANCE model to estimate the lifetime costs, life-years, and QALYs of the two treatment pathways displayed in Table 3. Efficacy estimates were obtained from pivotal clinical trials for each regimen.

Table 4. Regimen Information

Regimen	Probability of Achieving HIV RNA < 50 Copies/mL at 48 Weeks (as Treated[a])	Increase in CD4 Cell Count at 48 Weeks (as Treated[a])	Annual Probability of Virologic Failure[b] After 6 Months (as Treated[a])	Clinical Trial
EFV + ABC/3TC	88.0%	211	5%	CNA30024[15]
EFV + TDF/FTC	79.5%	190	5%	Gilead 934[16]
LPV/r + TDF/FTC	99.0%	185	5%	M02-418[17]
LPV/r + 3TC/ZDV	97.6%	159	5%	MONARK[18]

HIV = human immunodeficiency virus; RNA = ribonucleic acid.

[a] The ADVANCE model utilizes efficacy information based on "as treated" populations rather than "intent to treat" populations because adverse events and their associated outcomes are considered separately in the model.

[b] We assumed all regimens had a 5% annual probability of virologic failure.

- No adverse events were modeled within ADVANCE with the exception of TDF-induced nephrotoxicity manifested as acute renal failure (Table 5).

Table 5. Acute Renal Failure

ADVANCE Input	Value
Annual rate	0.87%[19]
90-day cost[a]	$0.00[b]
Utility decrement	–0.320[20]
Probability of swap given ARF	100.00%

ARF = acute renal failure.

[a] All currency expressed in US dollars.

[b] Based on a lack of reliable data, a conservative assumption of $0.00 was used for the cost of ARF.

Cost-effectiveness Analysis

- Outputs from the ADVANCE simulation model (Table 6) were entered as inputs to the HLA-B*5701 cost-effectiveness model to generate lifetime costs, life years, and QALYs associated with each treatment strategy.

Table 6. ADVANCE Output Used as Inputs to the HLA-B*5701 Cost-effectiveness Model

ADVANCE Scenario (First Line Second Line)	Lifetime Total Costs (Mean, 95% CI)[a]	Life-Years (Mean, 95% CI)	QALYs (Mean, 95% CI)
EFV + ABC/3TC followed by LPV/r + TDF/FTC	$306,279.43 ($302,591.89, $309,966.96)	15.44 (15.31, 15.57)	12.48 (12.38, 12.59)
EFV + TDF/FTC followed by LPV/r + 3TC/ZDV	$309,865.24 ($306,155.58, $313,574.89)	15.45 (15.32, 15.58)	12.46 (12.35, 12.56)

CI = confidence interval

Costs and outcomes were discounted at 3% per annum. All costs are reported in 2005 US dollars.

Results

Table 7. ABC/3TC + EFV With Prospective Testing Versus TDF/FTC + EFV

Outcome	ABC/3TC + EFV With Prospective Testing	TDF/FTC + EFV	Incremental
Total costs (per patient)[a]	$309,402	$312,796	–$3,394
Mean life expectancy (years)	15.61	15.62	–0.01
QALYs	12.62	12.59	0.03
Incremental cost per QALY gained[a,b]			ABC/3TC + EFV with prospective testing dominates TDF/FTC + EFV (–$129,329)

[a] All currency expressed in United States dollars.

[b] Difference in total costs divided by difference in total QALYs

- HLA-B*5701 testing and conditional initiation of ABC/3TC + EFV is a dominant strategy compared with initiation of TDF/FTC + EFV.
- The HLA-B*5701 cost-effectiveness model was sensitive to the lifetime costs and QALYs of the two treatment pathways, the cost of HLA-B*5701 testing, and the negative predictive value of HLA-B*5701 testing.
- Results were relatively insensitive to changes in all other parameters.

Discussion

- This analysis suggests that the use of ABC/3TC, as part of initial HAART and guided by HLA-B*5701 testing, provides similar quality-adjusted life expectancy at a reduced cost, relative to the use of TDF/FTC.
- This research differed from earlier work on this topic in that HLA-B*5701 testing was evaluated in real-world, clinical practice setting as compared to a blinded clinical trial protocol.
 - The true value of prospective testing is the information provided to the patient and the clinician and the differences in patient management resulting from known HLA-B*5701 status.
 - Evaluating the use of HLA-B*5701 testing in a real-world, clinical practice setting results in a small improvement in the cost-effectiveness ratio compared to when the patient and prescribing clinician are blinded to test results.
 - The analysis does not explicity consider qualitative benefits associated with testing such as increased patient and physician confidence during early ABC treatment nor does the analysis reflect the detriment of the loss of a potentially viable antiretroviral regimen when HSR is misdiagnosed. Inclusion of these benefits, while difficult to quantify, would further enhance the cost-effectiveness of ABC/3TC.

Conclusion

- Conditional initiation of ABC/3TC + EFV is favored over TDF/FTC + EFV as first-line choice of HAART.
- HLA-B*5701 testing and conditional initiation of ABC/3TC + EFV is a less costly and more effective treatment strategy compared with initiation of TDF/FTC + EFV.

Acknowledgements

The authors would like to extend their thanks to Dr. Benjamin Young, Dr. Trevor Hawkins, Dr. Princy Kumar, Dr. Edwin DeJesus, Dr. Charles Hicks, and Mr. Michael Chambers for their invaluable contributions to this study.

References

1. Mallal S, Phillips E, Carosi G, Molina JM, Workman C, Tomazic J, et al; for the PREDICT-1 Study Team.HLA-B*5701 screening for hypersensitivity to abacavir. N Engl J Med. 2008 Feb 7;358(6):568-79.
2. Kauf TL, Saag MS, Kitahata M, Gazzard B, Earnshaw SR, Graham CN, et al. Simulation Modeling of Adherence and Resistance on Long-Term Outcomes in HIV. 4th Annual Meeting of the Infectious Diseases Society of America, October 12-15 2006, Toronto, ON, Canada
3. Red Book for Windows, Version 61129, Volume 44. Montvale: Thomson PDR; April 2007.
4. LabCorp. HLA test. 2007. Available at: http://www.labcorp.com/datasets/labcorp/html/chapter/mono/ hl002200.htm. Accessed July 17, 2007.
5. US Department of Health and Human Services. Guidelines for the use of antiretroviral agents in HIV-1-infected adults and adolescents. Developed by the DHHS Panel on Antiretroviral Guidelines for Adults and Adolescents (OARAC). October 10, 2006. 2007. Available at: http://www.aidsinfo.nih.gov/ContentFiles/AdultandAdolescentGL.pdf. Accessed July 7, 2008.
6. RTI Health Solutions (RTI-HS). Clinician interviews. Data on file. May 2007.
7. Rauch A, Nolan D, Martin A, McKinnon E, Almeida C, Mallal S. Prospective genetic screening decreases the incidence of abacavir hypersensitivity reactions in the Western Australian HIV cohort study. Clin Infect Dis 2006;43(1):99-102.
8. Reeves I, Fisher M, Churchill D. Clinical utility of HLA-B*5701 testing in a UK clinic cohort. Abstract presented at the 13th Conference on Retroviruses and Opportunistic Infections; Denver, CO. Feb 5-8, 2006. Abstract No. 667a.
9. Waters L, Gritz A, Maitland D, Nelson M. HLA-B5701 testing and abacavir hypersensitivity: a single centre experience. Abstract presented at the 8th International Congress on Drug Therapy in HIV Infection; Glasgow, United Kingdom. Nov 12-16, 2006. Abstract No. PL9.2.
10. Zucman D, Stegman S, Caillat-Zucman S. Prospective screening for human leukocyte antigen B*5701 avoids abacavir hypersensitivity reaction in the ethnically mixed French HIV population. J AIDS 2007;45(1):1-3.
11. EPZICOM. [Package insert.]. Research Triangle Park, NC: GlaxoSmithKline. 2007
12. Kauf TL, Roskell N, Shearer A, Gazzard B, Mauskopf J, Davis EA, et al. A Predictive Model of Health State Utilities for HIV Patients in the Modern Era of Highly Active Antiretroviral Therapy. Value Health. 2008 May 20. [Epub ahead of print]
13. Ingenix.The Essential RBRVS. A Comprehensive listing of RBRVS values for CPT and HCPCS codes. Ingenix Inc: 2007.
14. RTI Health Solutions (RTI-HS). Ad hoc analysis of Integrated Health Care Information services among likely hypersensitivity reaction patients. June 2007.
15. DeJesus E, Herrera G, Teofilo E, Gerstoft J, Buendia CB, Brand JD, et al, for the CNA30024 Study Team. Abacavir versus zidovudine combined with lamivudine and efavirenz, for the treatment of antiretroviral-naive HIV-infected adults. Clin Infect Dis 2004;39:1038-46.
16. Gallant JE, DeJesus E, Arribas JR, et al. Tenofovir DF, emtricitabine, and efavirenz vs. zidovudine, lamivudine, and efavirenz for HIV. N Engl J Med 2006;354(3):251-60.
17. Johnson MA, Gathe JC Jr, Podzamczer D, et al. A once-daily lopinavir/ritonavir-based regimen provides noninferior antiviral activity compared with a twice-daily regimen. J Acquir ImmuneDefic Syndr 2006;43(2):153-60.
18. Delfraissy J-F, Flandre P, Delaugerre C, Horban A. MONARK Trial (monotherapy antiretroviral kaletra): 48-week analysis of lopinavir/ritonavir (LPV/r) monotherapy compared to LPV/r + zidovudine/lamivudine (AZT/3TC) in antiretroviral-naïve patients. Abstract presented at the International AIDS Conference, Toronto, Ontario, Canada. Aug 13-18 2006. Abstract No. ThLB0202.
19. Nelson MR, Katlama C, Montaner JS, Cooper DA; Gazzard B, Clotet B, et al., for the Tenofovir DF Expanded Access Team. The safety of tenofovir disoproxil fumarate for the treatment of HIV infection in adults: the first 4 years. AIDS 2007;21:1273-81.
20. Bell CM, Chapman RH, Stone PW, Sandberg EA, Neumann PJ. An off-the-shelf help list: a comprehensive catalog of preference scores from published cost utility analyses. Boston: Program on the Economic Evaluation of Medical Technology, Harvard Center for Risk Analysis, Harvard School of Public Health. US Med Decis Mak 2001;21(4):288-94.

AIDS 2006 – 18th International AIDS Conference, 3–8 August 2008, México City, México