CROI 2010
17th Conference on Retroviruses and Opportunistic Infections
February 16-19, 2010
San Francisco, CA
Abstract K-128
Poster # 515

Sustained Antiretroviral Effect of Raltegravir at Week 156 in the BENCHMRK Studies, and Exploratory Analysis of Late Outcomes based on Early Virologic Responses

J. J. Eron[1], D. A. Cooper[2], R. T. Steigbigel[3], B. Clotet[4], H. Wan[5], A. Meibohm[5], P. Sklar[5], B-Y. Nguyen[5], and H. Teppler[5] for the BENCHMRK-1 and 2 Study Groups
[1]University of North Carolina, USA; [2]University of New South Wales, Sydney, Australia; [3]SUNY at Stony Brook, USA; [4]University of Barcelona, Spain; [5]Merck Research Laboratories, North Wales, PA, USA

Joseph J. Eron, MD
UNC School of Medicine
Division of Infectious Diseases
Chapel Hill, NC 27599
joseph_eron@unc.edu

Abstract

Background: BENCHMRK (BM-) 1 and 2 are ongoing Phase III studies of raltegravir (RAL) 400 mg bid plus optimized background therapy (OBT) versus placebo (PBO) plus OBT. Week 156 efficacy results are reported, and an exploratory analysis evaluating late (week 96) efficacy outcomes of patients (pts) with low level viremia (LLV) between study week (wk) 16 and 48 compared to pts with continuous suppression is provided.

Methods: Pts failing ART with 3 class resistant HIV were randomized to RAL + OBT or PBO + OBT in a blinded comparison. Pre-specified efficacy endpoints were vRNA <50 and <400 copies/mL (c/mL) and mean change from baseline in CD4 counts. Studies were analyzed separately and combined. Exploratory analysis of wk 96 outcomes (% <50 c/mL) was conducted in RAL pts from both studies categorized by vRNA levels between wk 16 and wk 48 into one of 3 groups: 1) continuous suppression, CS (<50 c/mL at all timepoints); 2) LLV (<400 c/mL at all timepoints with at least one timepoint >50 c/mL); 3) not suppressed, NS (>400 c/mL at one or more timepoints).

Results: Baseline characteristics were comparable; overall, median baseline CD4 counts were 105 and 132, and mean vRNA log10 c/mL were 4.7 and 4.7 for RAL and PBO, respectively. BM-2 results (see table) show RAL maintaining ~30% treatment benefit and greater CD4 increase vs PBO. vRNA was <400 c/mL in 50% and 21% of RAL vs PBO pts at wk 156.

	% Pts with vRNA < 50 c/mL (95% CI), NC=F*			CD4 Change from Baseline (cells/µL) (95% CI), OF*		
	Wk 48	Wk 96	Wk 156	Wk 48	Wk 96	Wk 156
RAL	60 (53, 66)	56 (49, 62)	47 (41, 54)	97 (83, 112)	117 (98, 136)	158 (131, 184)
PBO	34 (26, 44)	23 (16, 31)	18 (11, 26)	39 (23, 55)	45 (24, 67)	55 (30, 80)

* NC=F: Non-completer = failure approach; OF: Observed Failure approach

Preliminary wk 156 results for BM-1 show similar trends: 53% and 25% <50 c/mL, 57% and 25% <400 c/mL and CD4 increases of 170 and 71 cells/µL for RAL and PBO, respectively. Results of exploratory analysis (OF approach) are shown below.

Category	Definition based on RNA c/mL between Wk 16-48 (N)	Outcome: % RNA< 50 c/mL Wk 48	Wk 96
Continuous suppression	<50 always (199)	100% (by definition)	90%
Low level viremia	50-400 at least once (111)	71%	72%
Not suppressed	>400 at least once (63)	17%	32%

Conclusion: In patients failing ART with 3-class resistant HIV, RAL efficacy was sustained through wk 156. The majority of patients with LLV before wk 48 had suppression at wk 96. Early LLV is not frequently associated with later overt virologic failure.

Introduction

Raltegravir (MK-0518, ISENTRESS™), an HIV-1 integrase strand-transfer inhibitor (InSTI)

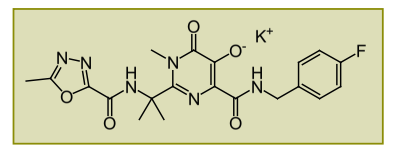

- Clinical efficacy: % of patients achieving HIV RNA < 50 copies/mL at Week 96
 - In ART-naive patients (in combination with TDF/3TC or TDF/FTC):
 - 83% in a phase II dose-ranging study[1]
 - 81% in STARTMRK (a phase III study)[2]
 - In patients failing therapy with triple class resistant virus (in combination with OBT):
 - 48% in a phase II dose-ranging study[3]
 - 57% in the combined BENCHMRK-1 and -2 (phase III) studies[4]

Overall Analysis, Week 156

BENCHMRK-1 & -2 Study Design

- Randomized, double-blind, placebo-controlled with DSMB
- Primary analysis at Week 16; secondary analysis at Week 48; planned study duration of **240** weeks
- Current analysis is Week 156 - the final double-blind comparison

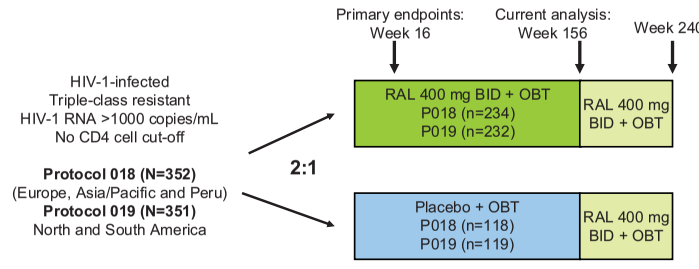
Primary endpoints: Week 16
Current analysis: Week 156
Week 240
HIV-1-infected
Triple-class resistant
HIV-1 RNA >1000 copies/mL
No CD4 cell cut-off
Protocol 018 (N=352)
(Europe, Asia/Pacific and Peru)
Protocol 019 (N=351)
North and South America
2:1
RAL 400 mg BID + OBT
P018 (n=234)
P019 (n=232)
RAL 400 mg BID + OBT
Placebo + OBT
P018 (n=118)
P019 (n=119)
RAL 400 mg BID + OBT

- After Week 156, double-blind phase was completed and all patients were offered open-label RAL until Week 240.

Statistical Methods

Efficacy Analyses

- Pre-defined endpoints examined at Week 156:
 - HIV RNA <50 copies/mL, <400 copies/mL
 - Non-Completer = Failure (NC=F) approach used for proportions over time analysis
 - Change from baseline in CD4 cell count
 - Observed Failure (OF) approach - baseline value carried forward for discontinuation due to lack of efficacy

Safety Analyses

- Clinical adverse events and lab abnormalities – displayed in 2 ways since ~ 68% greater duration of double-blind therapy for RAL vs placebo (mean 119 vs 71 weeks) due to more discontinuations from placebo due to virologic failure
 - percent of patients (n/N) with event
 - exposure-adjusted event rates (number of patients with event /100 person-years (PYR) exposure)

Results

Table 1. Patient Disposition

	Raltegravir + OBT (N = 462)		Placebo + OBT (N = 237)	
	N	(%)	N	(%)
Completed 156 wk Double-Blind Phase – continuing to Open label RAL	247	(53)	48	(20)
Entered Open-Label post VF* phase	99	(21)	131	(55)
Discontinued study	116	(25)	58	(24)
-- Discontinued due to adverse event	22	(5)	13	(5)

* Definition of virologic failure:
1) <1 $\log_{10}$ ↓ HIV RNA from baseline and HIV RNA >400 copies/mL at week 16, OR
2) virologic relapse: >1 $\log_{10}$ ↑ HIV RNA above nadir or >400 copies/mL from nadir after response <400 copies/mL (on 2 consecutive measurements at least 1 week apart).
After week 48, virologic failure is defined as: confirmed HIV RNA >50 copies/mL on 2 consecutive measurements at least 1 week apart.

Table 2. Summary of Adverse Events
Percent of Patients (Exposure Adjusted Rate†)

	Raltegravir + OBT (N = 462)		Placebo + OBT (N = 237)	
Patient-years (PYR) at risk	1051		322	
	%	(rate†)	%	(rate†)
Clinical Adverse Events	93.7	(41.2)	89.9	(66.1)
Drug-related‡	59.5	(26.2)	59.9	(44.1)
Serious	29.0	(12.7)	22.8	(16.8)
Serious & drug-related	3.0	(1.3)	3.8	(2.8)
Deaths	3.5	(1.5)	3.4	(2.5)
Patient discontinued	4.3	(1.9)	5.5	(4.0)
Laboratory Adverse Events	34.6	(15.2)	27.4	(20.2)
Drug-related‡	19.0	(8.4)	15.2	(11.2)
Serious	0.9	(0.4)	0.4	(0.3)
Serious & drug-related	0.2	(0.1)	0.0	(0.0)
Patient discontinued	0.2	(0.1)	0.0	(0.0)

† per 100 person-years (PYR).
‡ Determined by the investigator to be possibly, probably, or definitely related to raltegravir or placebo (alone or in combination with OBT).

Percent of Patients (95% CI) with HIV RNA <50 Copies/mL (NC=F Approach)

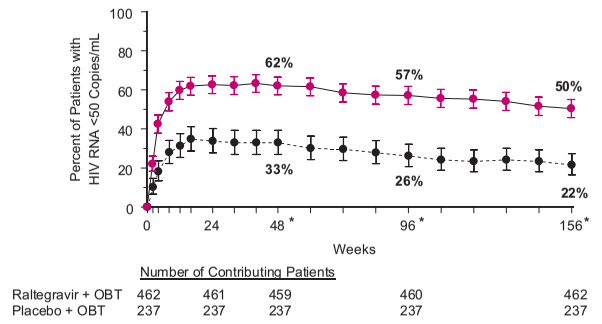
Percent of Patients with HIV RNA <50 Copies/mL
62%
57%
50%
33%
26%
22%
Weeks
Number of Contributing Patients
Raltegravir + OBT 462 461 459 460 462
Placebo + OBT 237 237 237 237 237

* p-value< 0.001. p-value was derived from a logistic regression model adjusted for baseline HIV RNA level ($\log_{10}$), first enfuvirtide use in OBT, first darunavir use in OBT, active PI in OBT.

Percent of Patients (95% CI) with HIV RNA <400 Copies/mL (NC=F Approach)

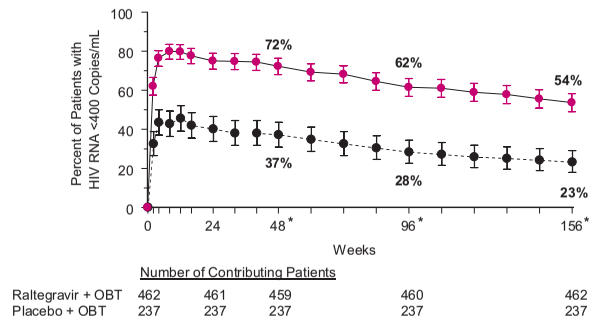
Percent of Patients with HIV RNA <400 Copies/mL
72%
62%
54%
37%
28%
23%
Weeks
Number of Contributing Patients
Raltegravir + OBT 462 461 459 460 462
Placebo + OBT 237 237 237 237 237

* p-value< 0.001. p-value was derived from a logistic regression model adjusted for baseline HIV RNA level ($\log_{10}$), first enfuvirtide use in OBT, first darunavir use in OBT, active PI in OBT.

Change From Baseline in CD4 Cell Count (cells/mm³) (OF Approach)

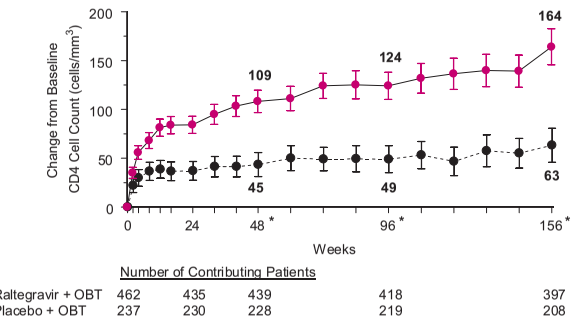
Change from Baseline CD4 Cell Count (cells/mm³)
109
124
164
45
49
63
Weeks
Number of Contributing Patients
Raltegravir + OBT 462 435 439 418 397
Placebo + OBT 237 230 228 219 208

* p-value< 0.001. For change from baseline in CD4 cell counts, p-value was derived from a mixed-effects model adjusted for: baseline CD4 cell count, stratum, treatment, visit, interactions between visit and previous variables.

References

1. Markowitz M, et al. *JAIDS* 2009;52(3):350-356.
2. Lennox J, et al. ICAAC 2009, abstract H-924b.
3. Gatell J, et al. *JAIDS* 2010, epub ahead of print.
4. Steigbigel R, et al. *Clin Infect Dis* 2010;50:605-612.

Exploratory Analysis by Early (wk 16-48) vRNA Response in RAL Treatment Group

Methods

- Inclusion criteria:
 - Patients who completed at least 48 weeks of double-blind treatment
- Early vRNA Response classification:
 - Continuous suppression; low level viremia; non-suppressed (intermittent)
 - Based on observed HIV RNA values between weeks 16 and 48 (see Table 3); 5 timepoints (wk 16, 24, 32, 40, 48)
- Missing data approach — Observed Failure (OF)
 - For HIV RNA, only discontinuations due to lack of efficacy counted as failures
 - For change in CD4 count, baseline value carried forward for discontinuations due to lack of efficacy

Table 3. Classification of Early vRNA Response

Category Name		Definition: observed vRNA (copies/mL) Week 16 – 48 (5 timepoints)	RAL N=462
Continuous Suppression	CS	All timepoints < 50	**199**
Low level Viremia	LLV	All < 400 with one or more > 50†	**111**
Not Suppressed	NS	intermittent >400 (not consecutive)	**63**
Not included	–	Pts who discontinued double blind treatment prior to wk 48‡	89

† of 5 possible time points between wk 16 and 48: 57% had one vRNA >50, 39% had 2-4 vRNA >50, and 4% had all values > 50 and < 400.
‡ 74% of discontinuations from RAL were due to virologic failure.

- Efficacy through week 156 by early vRNA response (CS, LLV and NS)
 - HIV RNA <50, <400 copies/mL
 - Change from baseline in CD4 cell count (cells/mm³)
- Association between early vRNA response category and prognostic factors (including rapid viral decay: defined as vRNA <50 at least once between wk 2-12)
 - Univariate Analysis
 - Multivariate Analysis
- Time to event analysis by early vRNA response category (CS and LLV)
 - Time to loss of virologic response, TLOVR (≥400 copies/mL)
 - Time to virologic response (<400 copies/mL)
- Integrase resistance-associated mutations (RAM) summarized by early vRNA response category (CS, LLV and NS) for available data

Results

Table 4. Baseline Characteristics by Early vRNA Response, RAL group

	CS N = 199	LLV N = 111	Intermittent NS N = 63
Plasma HIV RNA ($\log_{10}$ copies/mL)			
median (min, max)	4.4 (2.3 – 5.8)	4.9 (2.6 – 5.9)	5.0 (2.8 – 5.9)
> 100,000, n (%)	37 (18.6)	47 (42.3)	33 (52.4)
CD4 count (cells/µL)			
median (min, max)	166 (2-792)	101 (1-740)	69 (1-469)
≤ 50, n (%)	33 (16.6)	41 (36.9)	29 (46.0)
CD4%, median (min, max)	11 (0-46)	9 (0-30)	5 (0-26)
CD8 count, median (min, max)	887 (167-2605)	729 (46-2296)	642 (69-2530)
CD8%, median (min, max)	62 (26-89)	61 (11-86)	62 (21-83)
CD4/CD8 ratio, median (min, max)	0.18 (0.01-1.23)	0.14 (0.01-0.66)	0.07 (0.01-0.91)
PSS (%): 0	13	7	13
1	29	33	33
2 or more	54	56	48
GSS (%): 0	17	19	25
1	38	47	38
2 or more	44	33	33

Table 5. Patient Status at Week 156 by Early vRNA Response, RAL group

	CS N = 199	LLV N = 111	Intermittent NS N = 63
Completed double-blind phase, and continuing on open-label raltegravir	157 (78.9)	91 (82.0)	25 (39.7)
Discontinued study	35 (17.6)	10 (9.0)	22 (34.9)
Lack of efficacy	2 (1.0)	1 (0.9)	9 (14.3)
Adverse event	6 (3.0)	0 (0.0)	1 (1.6)
Other reason†	27 (13.6)	9 (8.1)	12 (19.0)
Virologic failure between wks 48-156, and entered Open-Label Post Virologic Failure (OLPVF) group	7 (3.5)	10 (9.0)	16 (25.4)

Data shown as number (%) of patients.
† includes consent withdrawn, lost to follow-up, did not enter extension, moved or relocated, clinical trial terminated at study site.

Percent of Patients in RAL Group with HIV RNA < 50 copies/mL (OF), by Early vRNA Response (wk 16-48)

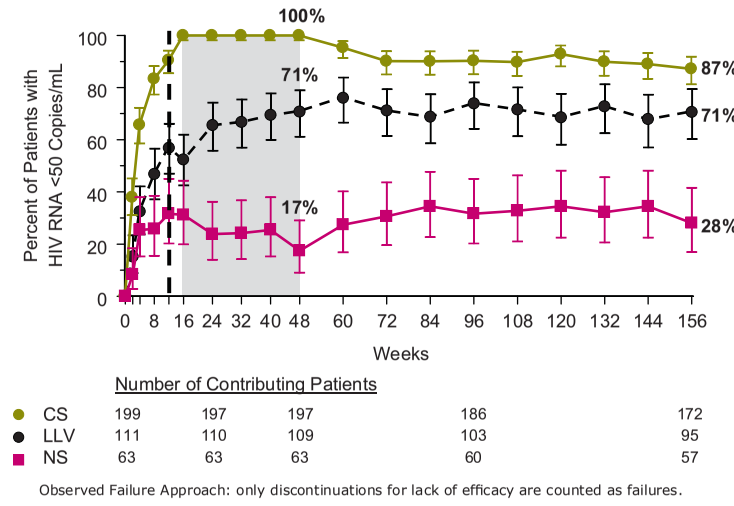
Percent of Patients with HIV RNA <50 Copies/mL
100%
71%
17%
87%
71%
28%
Weeks
Number of Contributing Patients
CS 199 197 197 186 172
LLV 111 110 109 103 95
NS 63 63 63 60 57

Observed Failure Approach: only discontinuations for lack of efficacy are counted as failures.

Percent of Patients in RAL Group with HIV RNA < 400 copies/mL (OF), by Early vRNA Response (wk 16-48)

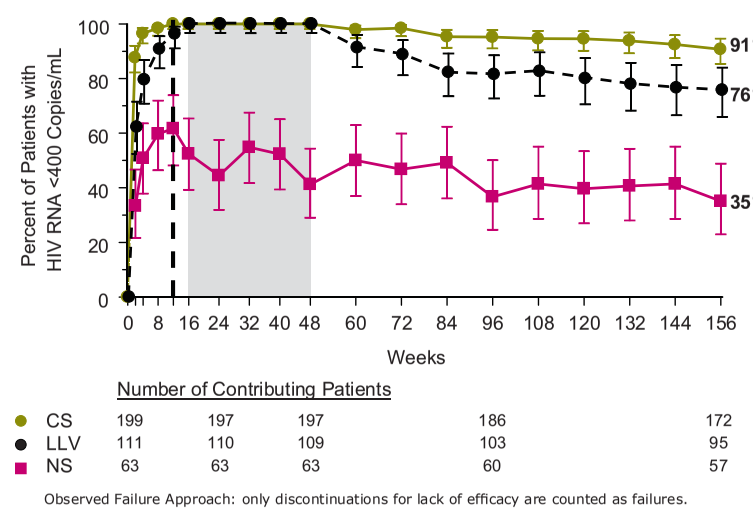
Percent of Patients with HIV RNA <400 Copies/mL
91%
76%
35%
Weeks
Number of Contributing Patients
CS 199 197 197 186 172
LLV 111 110 109 103 95
NS 63 63 63 60 57

Observed Failure Approach: only discontinuations for lack of efficacy are counted as failures.

Change from Baseline in CD4 Cell Count (cells/mm³) (OF) by Early vRNA Response, RAL Group (wk 16-48)

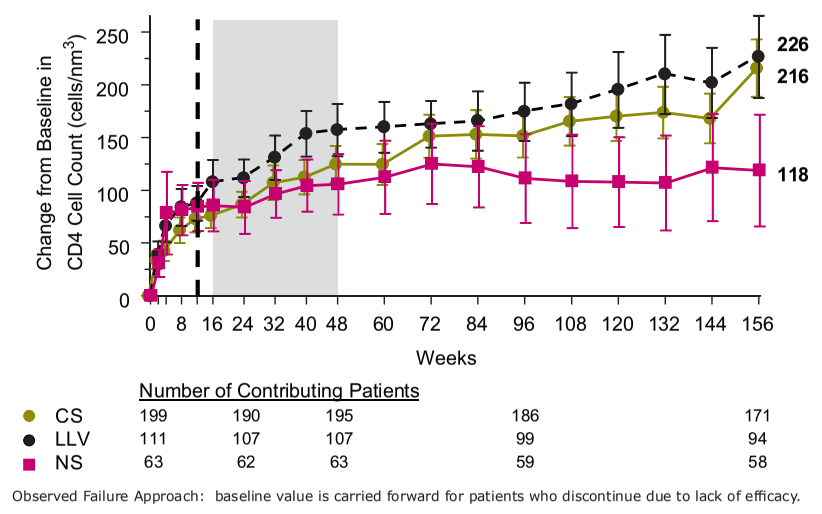
Change from Baseline in CD4 Cell Count (cells/mm³)
226
216
118
Weeks
Number of Contributing Patients
CS 199 190 195 186 171
LLV 111 107 107 99 94
NS 63 62 63 59 58

Observed Failure Approach: baseline value is carried forward for patients who discontinue due to lack of efficacy.

Table 6. Association Between Early vRNA Response Category (CS, LLV, NS) and Baseline Prognostic Factor (univariate: categorical variable)

Baseline Factor	P-value†
HIV RNA (≤100,000 : >100,000) copies/mL	<0.001
CD4 cell count (≤50 : >50 and ≤200 : >200) cells/µL	<0.001
Rapid viral decay (yes : no)‡	<0.001
GSS	0.215
PSS	0.997

† A k-by-k contingency table using each prognostic factor was conducted; p-value from Mantel-Haenszel chi-square test is provided.
‡ Rapid viral decay: HIV RNA <50 copies/mL at least once between wk 2-12.

- All factors except GSS/PSS appear correlated with early vRNA response category
- In a univariate analysis using continuous variables, baseline characteristics highly correlated with early vRNA response category included $\log_{10}$ HIV RNA, CD4 cell count, CD8 cell count and CD4/CD8 ratio (p<0.001 for all except CD8 [p=0.002]).

Multivariate Analysis

- In this analysis
 - CD4 and vRNA are continuous variables
 - GSS, PSS, and rapid viral decay are categorical variables
- Only rapid viral decay (vRNA < 50 cp/mL between wk 2-12) was significantly associated with early vRNA response category (p <0.001).

Table 7. Responders† at Week 96 and 156 in CS and LLV Groups

	Week 96	P-value‡	Week 156	P-value‡
CS group	182/199	0.004	175/199	0.009
n/N (%)	(91%)		(88%)	
LLV group	89/111		85/111	
n/N (%)	(80%)		(77%)	

† Responders: Patients who have not yet met protocol definition of Virologic Failure (confirmed HIV RNA > 50 cp/mL, on 2 consecutive measurements ≥1 week apart).
‡ for difference between CS and LLV groups.
CS = continuous suppression: vRNA < 50 cp/mL from wk 16 to 48.
LLV = low-level viremia: vRNA < 400 cp/mL from wk 16 to 48 but >50 at least once.

HIV Integrase Mutations in Patients with Virologic Failure

- Preliminary and partial data indicate approximately 35-55% of patients with virologic failure who have had resistance testing performed show one or more of the RAL signature mutations (AA 143, 148, or 155):
 - CS group: 6/11 (55%)
 - LLV group: 4/11 (36%)
 - NS group: 24/44 (55%)

Time to Loss of Virologic Response (TLOVR, ≥400cp/mL) by CS and LLV Groups

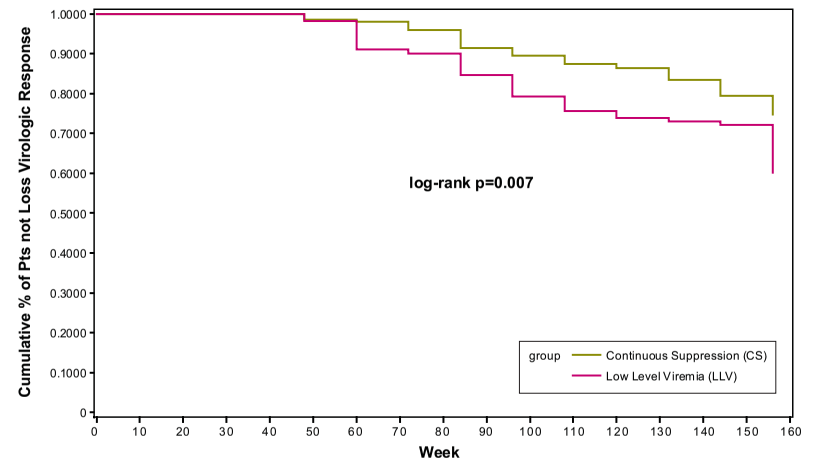
Cumulative % of Pts not Loss Virologic Response
log-rank p=0.007
group
Continuous Suppression (CS)
Low Level Viremia (LLV)
Week

- Time to Virologic Response (<400cp/mL) is significantly shorter in CS group compared to LLV group (log-rank p <0.001)

Conclusions

- In HIV-infected, treatment-experienced patients failing ART with triple-class resistance:
 - RAL 400 mg b.i.d. plus OBT, compared to PBO plus OBT, had potent, superior, and durable antiretroviral and immunological efficacy sustained through Week 156.
 - 50% in RAL group sustained vRNA < 50 cp/mL
 - RAL 400 mg b.i.d. plus OBT was generally well tolerated as compared to PBO plus OBT.
 - Few discontinuations due to adverse events
- For an exploratory analysis, patients were categorized by early virologic responses (continuous suppression, low level viremia, and intermittent non-suppressed) based on vRNA between wk 16-48
 - Patients with low level viremia (LLV, 50<vRNA<400 at least once) had more advanced baseline disease characteristics (RNA↑, CD4↓) compared with the continuous suppression (CS, all vRNA < 50) group.
- In exploratory analyses to examine correlation of early virologic response with baseline prognostic factors (including rapid viral decay, i.e., vRNA < 50 cp/mL at least once between wk 2-12)
 - Univariate analyses indicate correlation with vRNA, CD4 count and rapid viral decay
 - In multivariate analysis, rapid viral decay showed strongest association
- In exploratory analyses of wk 156 outcomes based on early virologic responses, patients with low level viremia demonstrated:
 - Favorable wk 156 virologic and immunologic outcomes
 - 71% < 50 cp/mL, 76% < 400 cp/mL
 - CD4 increase from BL +226 cells/mm³
 - Significantly shorter time to loss of virologic response (TLOVR ≥ 400 cp/mL) compared to CS group.

Acknowledgements

All patients who participated in the BENCHMRK-1 & 2.

BENCHMRK-1 Investigators: *Australia:* Allworth A, Anderson J, Bloch M, Cooper DA, Hoy J, Moore RJ, Workman C; *Belgium:* Clumeck N, Colebunders R, Florence E, Moutschen M; *Denmark:* Gerstoft J, Larsen C, Mathiesen L, Pedersen C; *France:* Delfraissy JF, Dellamonica P, Katlama C, Molina JM, Raffi F, Reynes J, Vittecoq D, Yeni P; *Germany:* Arasteh K, Fatkenheuer G, Jaeger H, Rockstroh J, Stoehr A; *Italy:* Aiuti F, Carosi G, Cauda R, Chiodo F, Di Perri G, Filice G, Galli M, Lazzarin A, Mazzarom I, Vullo V; *Peru:* Castaneda M, Florez A, Mendo F, Paredes A, Salazar R, Ticona E; *Portugal:* Antunes R, Diniz A, Mansinho K, Saraiva da Cunha J, Sarmento R, Teofilo E, Vera J; *Spain:* Arrizabalaga J, Clotet Sala B, Domingo Pedrol P, Gatell Artigas J, Moreno Guillen S, Soriano Vazquez V; *Switzerland:* Hirschel B, Opravil M, Weber R; *Taiwan:* Lin H-H, Sheng W-H, Wang J-H; *Thailand:* Sungkanuparph S, Suwanagool S.

BENCHMRK-2 Investigators: *Brazil:* Grinsztejn B, Madruga JV, Schechter M; *Canada:* Baril J-G, Loutfy, MR, Montaner JS, Tremblay C, Tsoukas CM, Vezina S; *Colombia:* Cortes JA, Mendoza H, Velez J; *Mexico:* Quintero Perez N, Ramos J, Rodriguez E; *Puerto Rico:* Morales-Ramirez JO, Sepulveda GE; *US:* Aberg J, Beatty GW, Benson P, Bolon RK, Bredeek UF, Bruno C, Campbell T, Campo R, Coodley GO, Corales RB, DeJesus E, Eron JJ, Fessel, WJ, Fetchick RJ, Gonzales CJ, Hicks C, Horberg MA, Klein DB, Kozal MJ, Kumar PN, LaMarca A, Lennox JL, Lichtenstein KA, Liporace R, Little SJ, Luetkemeyer A, Mariuz P, Markowitz M, McMahon DK, Perez G, Pierone G, Reichman RC, Rhame F, Shalit P, Short W, Skolnik PR, Steigbigel RT, Tedaldi EM, Ward DJ, Wiznia AA, Wright DP

Merck Research Laboratories: H. Wan, A. Rodgers, J. Zhao, X. Xu, D. Hazuda, M. Miller, R. Danovich, R. Rhodes, B. Jackson, K. Strohmaier, P. Sklar, R. Leavitt, H. Teppler, B-Y. Nguyen.

Special thanks to J. Zhao for additional exploratory analyses.

Copyright © 2010 Merck & Co., Inc., Whitehouse Station, New Jersey, USA, All Rights Reserved

2010-0101 CROI Feb-2010 Strohmaier